IDWeek 2022 | Poster #P1687

Antimicrobial Activity of Ceftibuten-Avibactam against a Global Collection of Enterobacterales from Patients with Complicated Urinary Tract Infections (2021)

Helio S. Sader, Cecilia G. Carvalhaes, Michael D. Huband, Rodrigo E. Mendes, and Mariana Castanheira

JMI Laboratories, North Liberty, IA, USA

Introduction

- Limited therapeutic options are currently available for the oral treatment of complicated urinary tract infections (cUTIs) caused by antimicrobial-resistant Enterobacterales.
- Ceftibuten is an oral cephalosporin active against Enterobacterales approved by the US FDA in 1995.
- Avibactam, a potent inhibitor of extended-spectrum β-lactamases (ESBLs), serine carbapenemases, and AmpC enzymes, can be administered orally.
- We evaluated the *in vitro* activity of ceftibuten-avibactam against Enterobacterales causing cUTI in medical centers worldwide.

Methods

- Organism Collection
 - Participant medical centers were invited to collect a specific number (25 to 60, depending on geographic region) of consecutive isolates (1/patient) from patients with cUTI in 2021.
 - Only the bacterial isolates that were the probable cause of a patient's infection were included in this investigation.
 - The organism collection included 3,216 isolates from 72 medical centers in 25 countries.
 - Isolates were mainly from the US (*n*=1,585; 29 centers) and Europe (*n*=1,410; 33 centers in 18 countries).
 - The organism collection also included *E. coli* isolates from Latin America (*n*=121; 6 centers in 5 countries) and Japan (*n*=100; 4 centers).
- Susceptibility Testing
 - Antimicrobial susceptibility was evaluated by the CLSI broth microdilution method.
 - Avibactam was present at a fixed concentration of 4 mg/L in combination with ceftibuten.
 - Current ceftibuten breakpoints published by CLSI (≤8 mg/L) and EUCAST (≤1 mg/L) were applied to ceftibuten-avibactam for comparison.
 - *E. coli*, *K. pneumoniae*, and *P. mirabilis* isolates were categorized as exhibiting an ESBL phenotype based on CLSI criteria; i.e., the isolate had an elevated MIC value (≥2 mg/L) for ceftazidime, ceftriaxone, or aztreonam.
 - Isolates were considered multidrug resistant (MDR) according to criteria defined in 2012 by the joint European and US Centers for Disease Control, which define MDR as nonsusceptible to ≥1 agent in ≥3 antimicrobial classes.
 - Carbapenem-resistant Enterobacterales (CRE) were defined as isolates that displayed imipenem or meropenem MIC values ≥4 mg/L; imipenem MIC results were not applied to *P. mirabilis* or indole-positive Proteeae due to their intrinsically elevated MIC values.

Results

- The most common organisms isolated in both the US and Europe were *E. coli*, *K. pneumoniae*, *P. mirabilis*, and indole-positive Proteeae (Figure 1).
- The most active agents against Enterobacterales were ceftibuten-avibactam (98.4%/99.6% inhibited at ≤1/≤8 mg/L), ceftazidime-avibactam (99.6% susceptible [S]), amikacin (99.1%S), and meropenem (98.2%S; Table 1 and Figure 2).
- Ceftibuten-avibactam ($MIC_{50/90}$, 0.03/0.06 mg/L) was 4-fold more potent than ceftazidime-avibactam ($MIC_{50/90}$, 0.12/0.25 mg/L) based on $MIC_{50/90}$ values.
- The most active oral agents were ceftibuten (89.3%S; 79.5% inhibited at ≤1 mg/L), levofloxacin (75.4%S), and trimethoprim-sulfamethoxazole (TMP-SMX; 73.4%S; Table 1 and Figure 2).
- Only ceftibuten-avibactam, ceftazidime-avibactam, meropenem, and amikacin were active against >90% of ESBL-phenotype isolates (Table 1 and Figure 3).
- Only ceftibuten-avibactam, ceftazidime-avibactam, and amikacin were active against >70% of CRE isolates (Table 1 and Figure 3).
- Ceftibuten-avibactam activity against CREs was similar to ceftazidime-avibactam ($MIC_{50/90}$, 2/>32 mg/L; 77.2% susceptible; Table 1 and Figure 3).
- CRE rates were 1.8%/1.9% in the US/EU; ceftibuten-avibactam inhibited 79.3%/81.5% of CRE isolates from the US/EU at ≤8 mg/L (72.4%/77.8% at ≤1 mg/L; data not shown).

Figure 1. Frequency of Enterobacterales causing complicated urinary tract infections in the United States (A) and Europe (B)

A. United States (*n*=1,585)

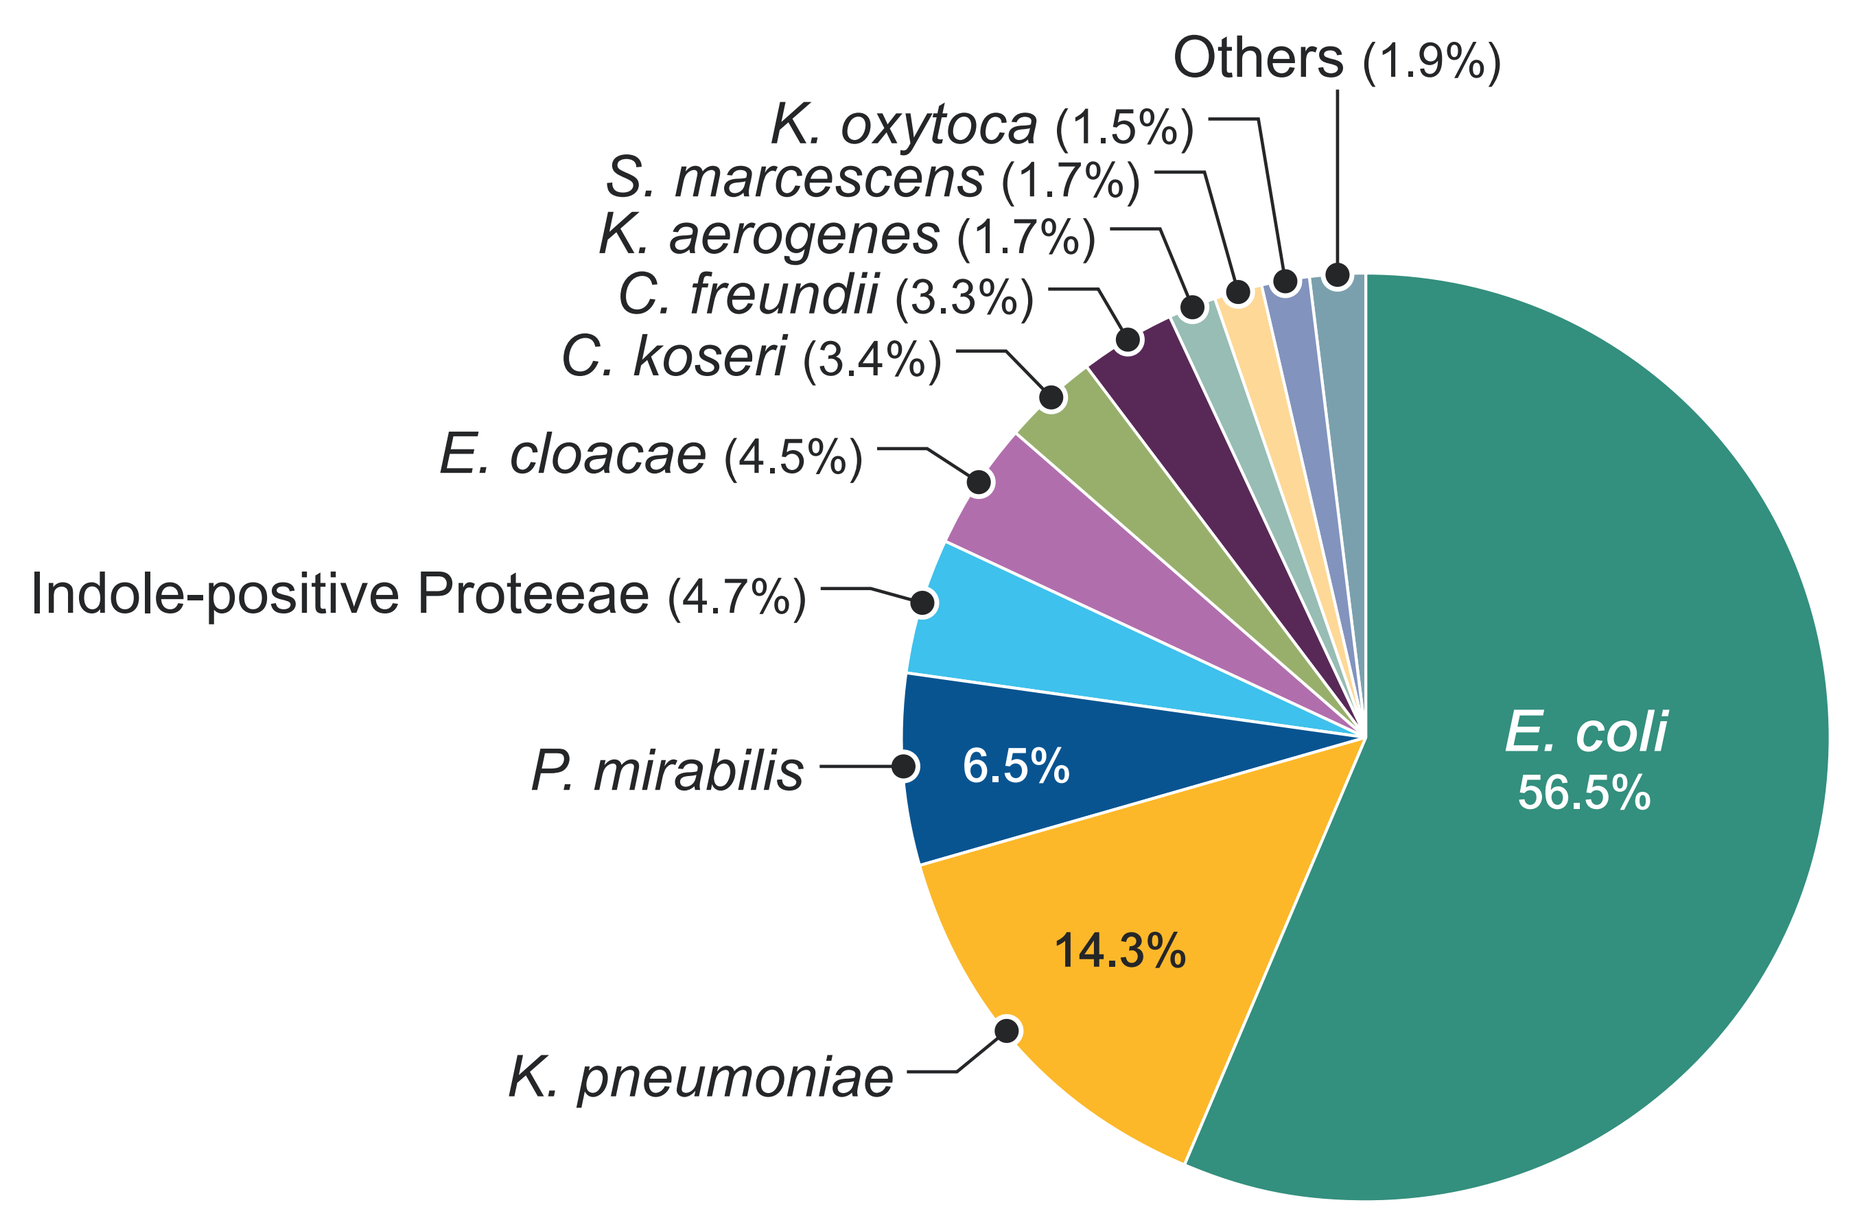

Others (1.9%)
K. oxytoca (1.5%)
S. marcescens (1.7%)
K. aerogenes (1.7%)
C. freundii (3.3%)
C. koseri (3.4%)
E. cloacae (4.5%)
Indole-positive Proteeae (4.7%)
P. mirabilis
6.5%
14.3%
K. pneumoniae
E. coli
56.5%

B. Europe (*n*=1,410)

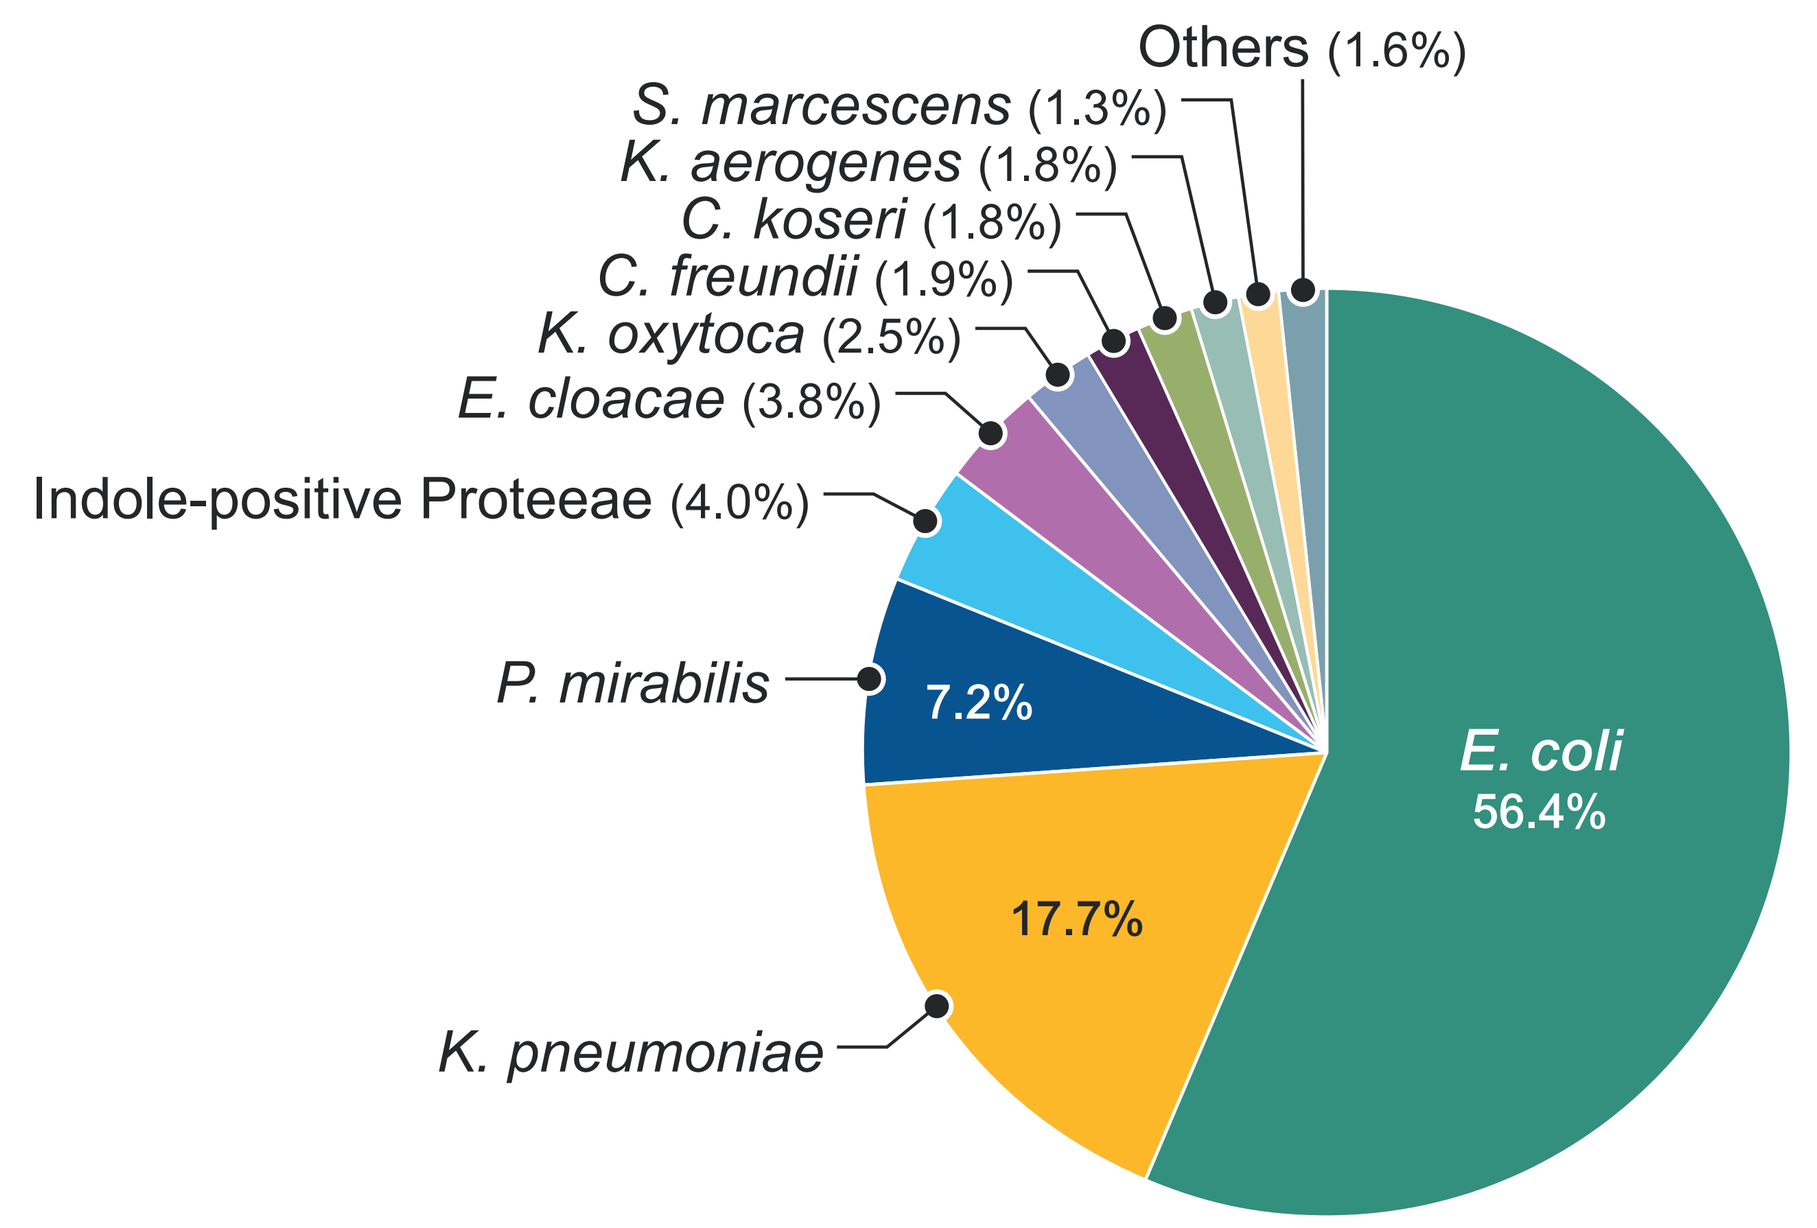

Others (1.6%)
S. marcescens (1.3%)
K. aerogenes (1.8%)
C. koseri (1.8%)
C. freundii (1.9%)
K. oxytoca (2.5%)
E. cloacae (3.8%)
Indole-positive Proteeae (4.0%)
P. mirabilis
7.2%
17.7%
K. pneumoniae
E. coli
56.4%

Figure 2. Antimicrobial susceptibility of Enterobacterales isolated from patients with complicated urinary tract infections

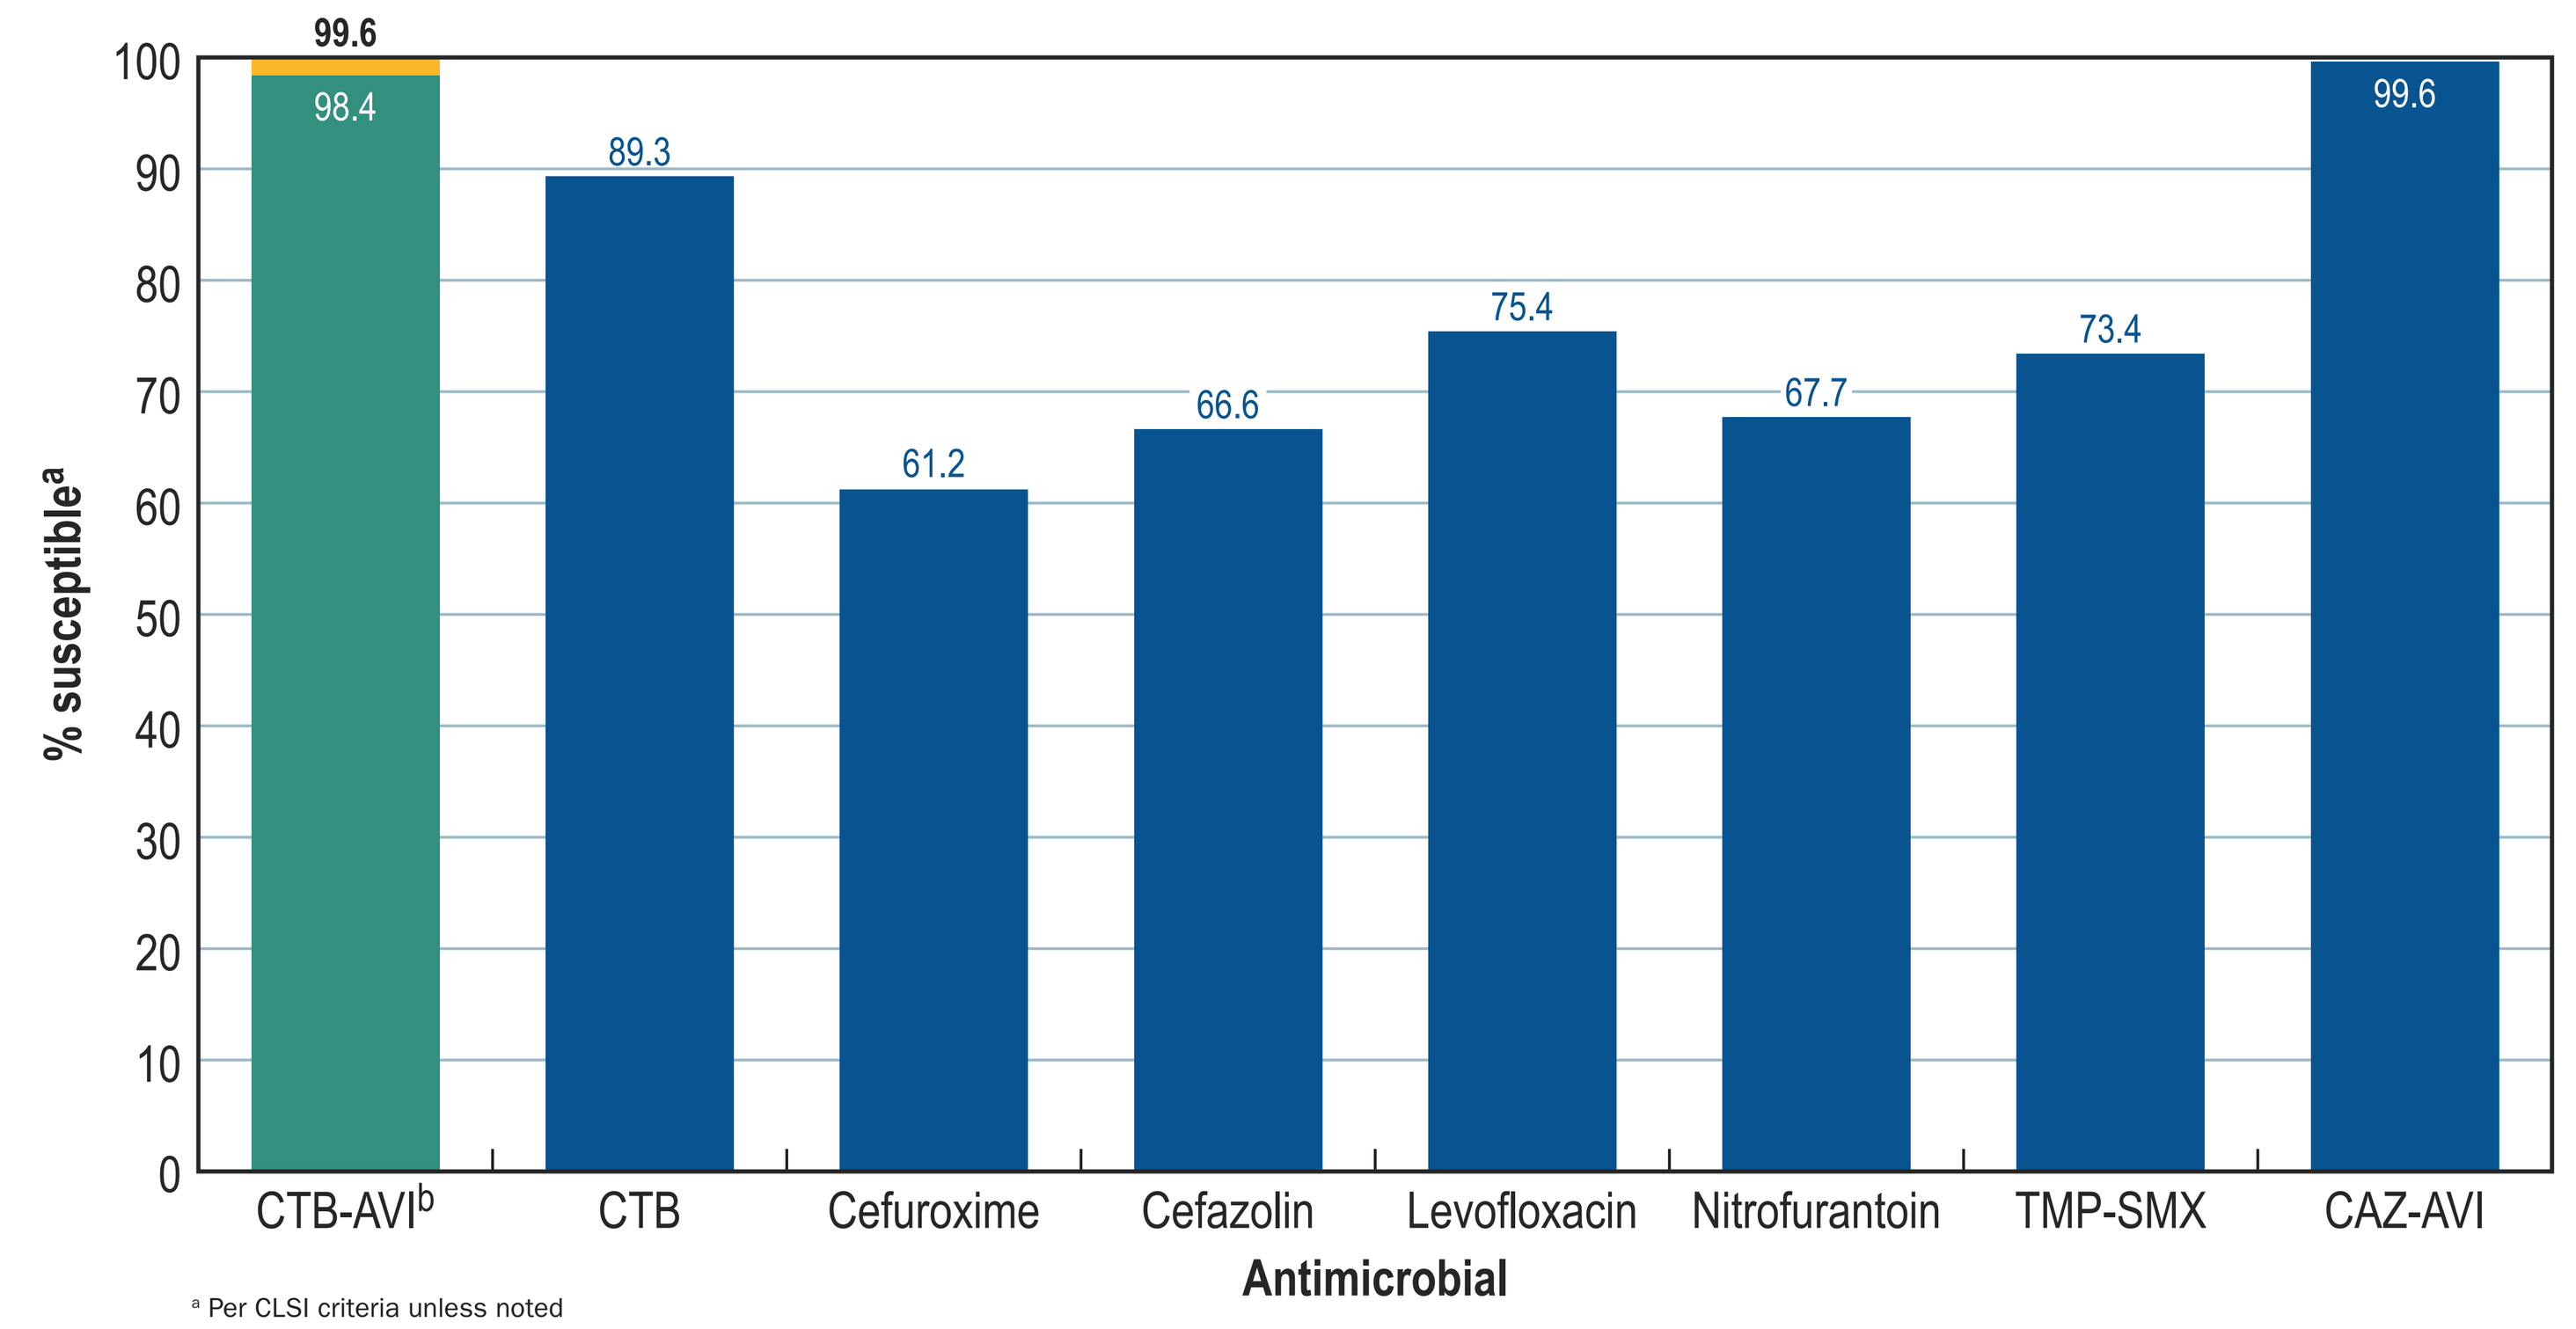

% susceptible[a]
99.6
98.4
89.3
61.2
66.6
75.4
67.7
73.4
99.6
CTB-AVI[b]
CTB
Cefuroxime
Cefazolin
Levofloxacin
Nitrofurantoin
TMP-SMX
CAZ-AVI
Antimicrobial

[a] Per CLSI criteria unless noted
[b] Percentage inhibited at ≤1 mg/L (green) and ≤8 mg/L (yellow).
Abbreviations: CTB-AVI, ceftibuten-avibactam; CTB, ceftibuten; TMP-SMX, trimethoprim-sulfamethoxazole; CAZ-AVI, ceftazidime-avibactam.

Figure 3. Activity of ceftibuten-avibactam against Enterobacterales-resistant subsets if isolates were collected from patients with complicated urinary tract infections

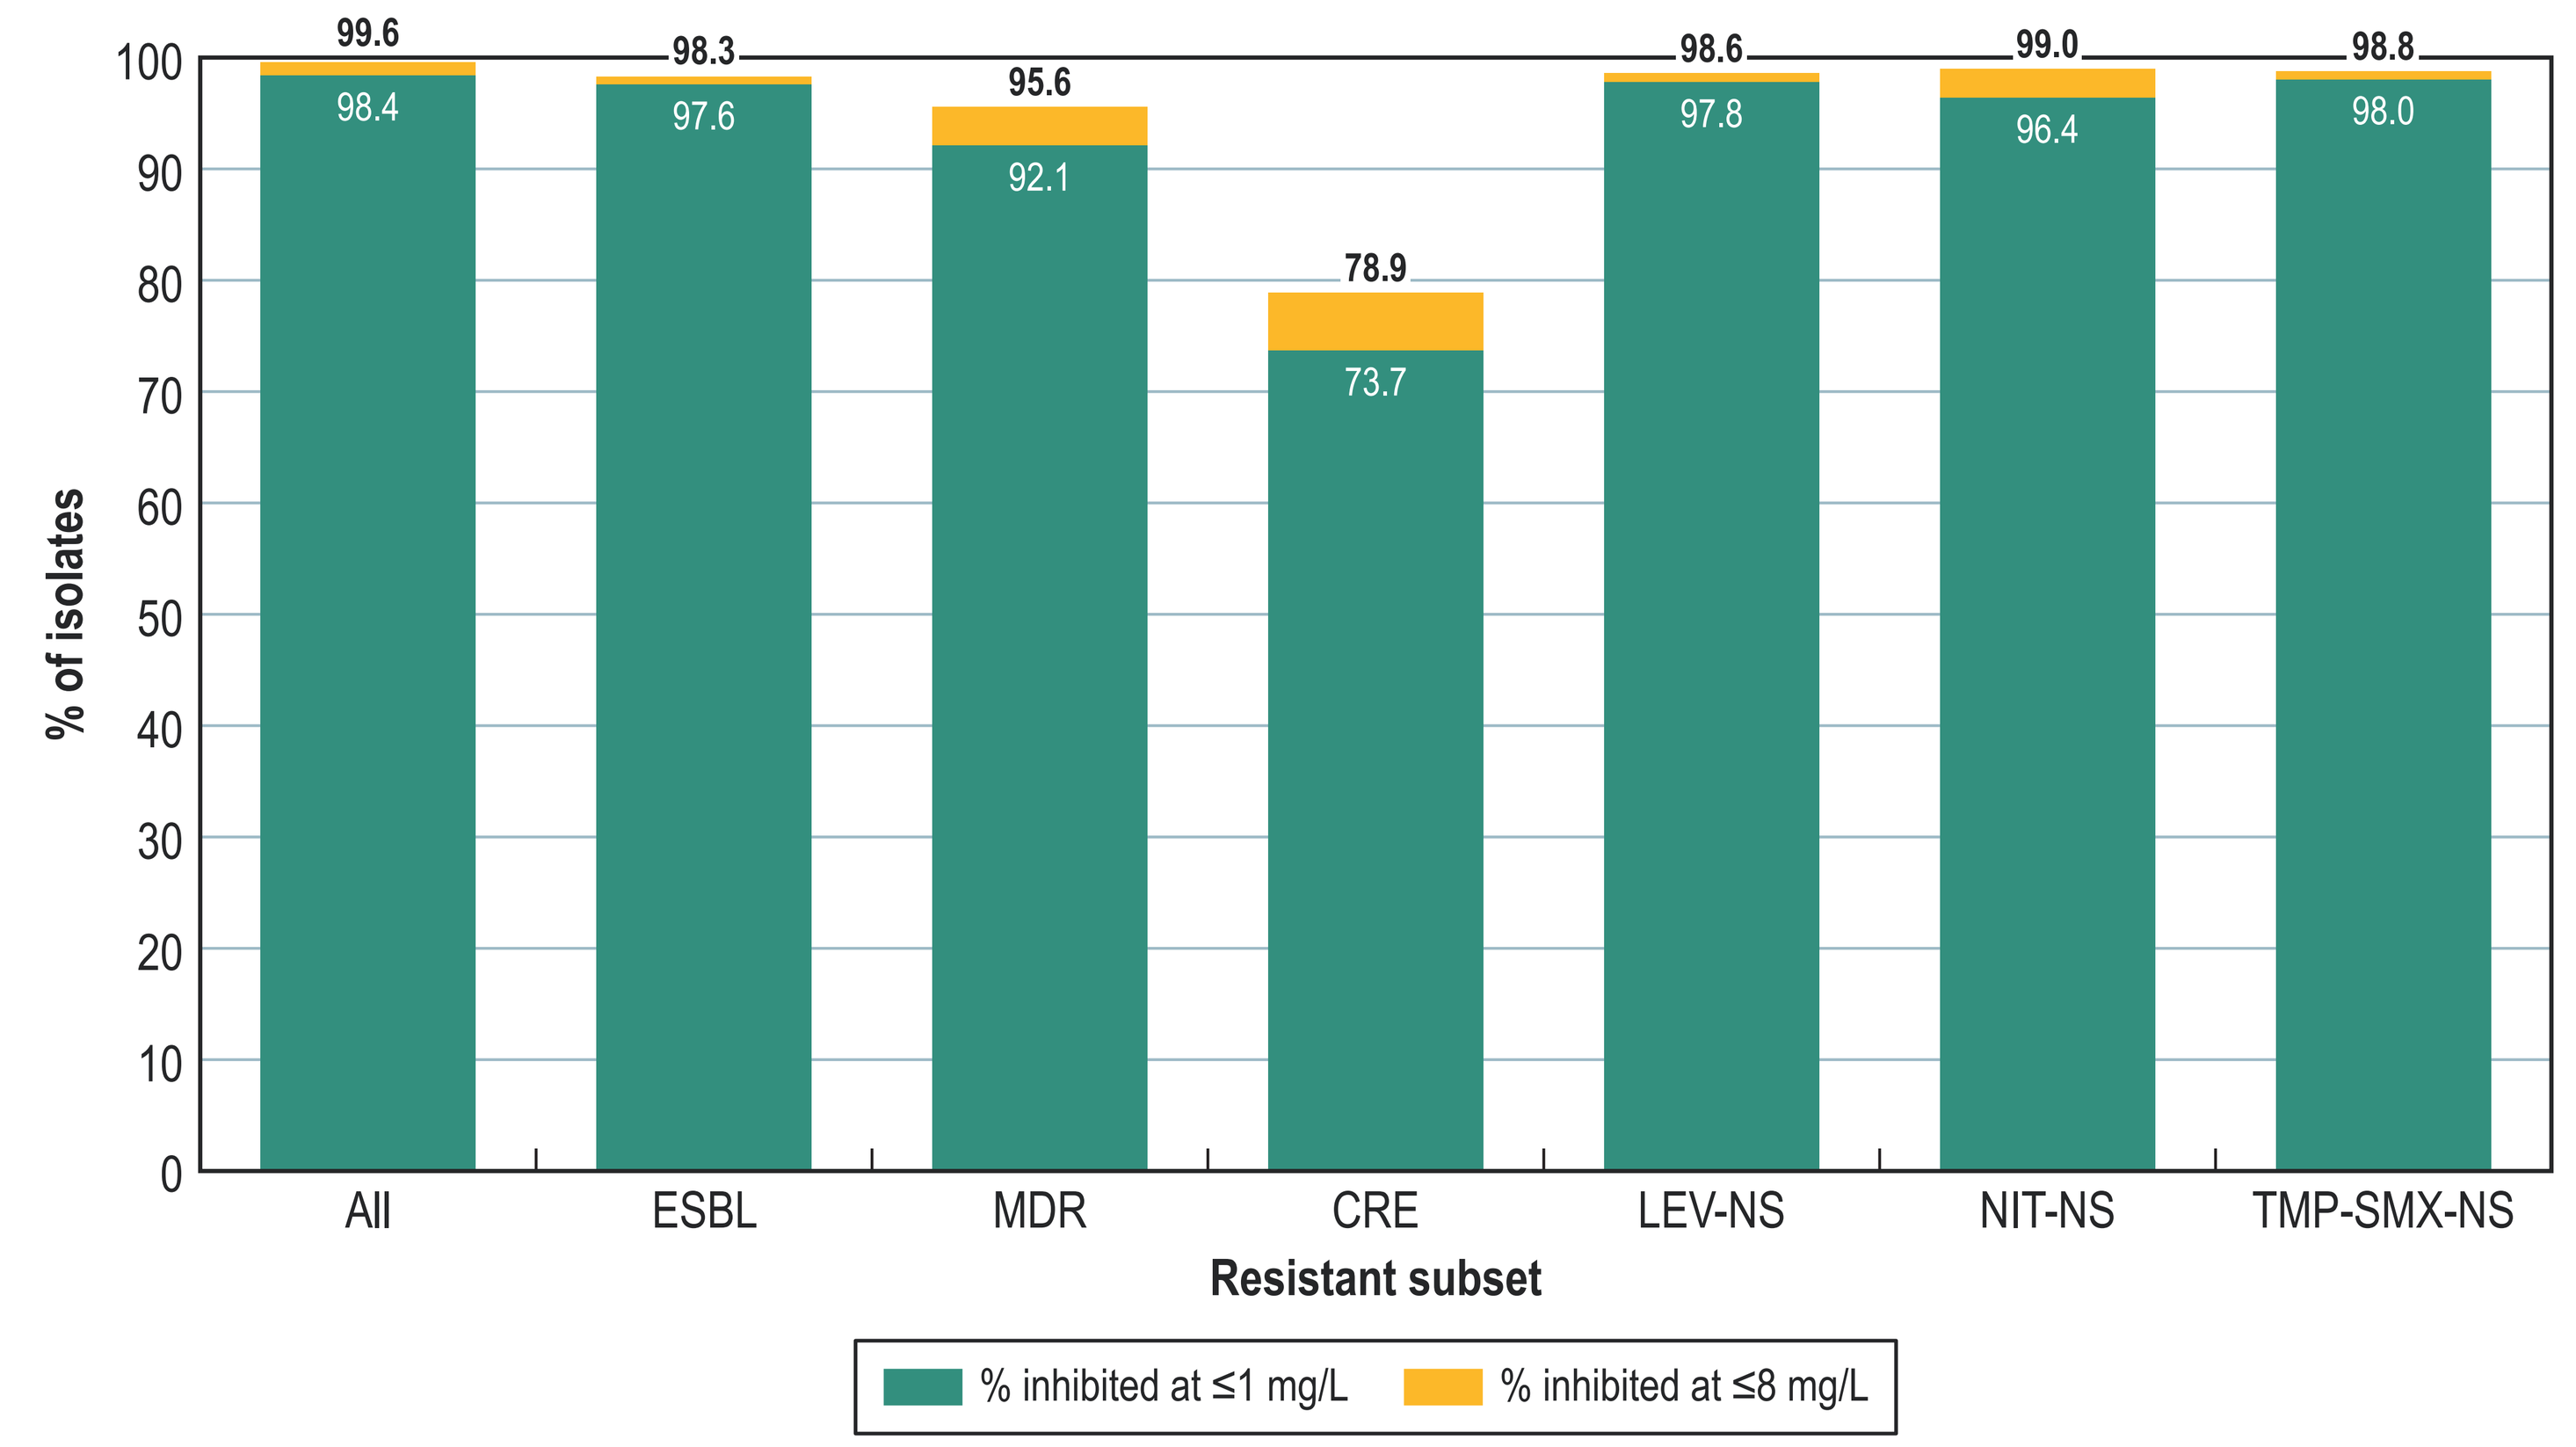

% of isolates
99.6
98.4
98.3
97.6
95.6
92.1
78.9
73.7
98.6
97.8
99.0
96.4
98.8
98.0
All
ESBL
MDR
CRE
LEV-NS
NIT-NS
TMP-SMX-NS
Resistant subset
% inhibited at ≤1 mg/L
% inhibited at ≤8 mg/L

Abbreviations: ESBL, extended-spectrum β-lactamase; MDR, multidrug-resistant; CRE, carbapenem-resistant Enterobacterales; LEV, levofloxacin; NS, nonsusceptible per CLSI criteria; NIT, nitrofurantoin; TMP-SMX, trimethoprim-sulfamethoxazole.

Table 1. Antimicrobial activity of ceftibuten-avibactam in comparison to oral and intravenous comparator agents tested against Enterobacterales

	% Susceptible per CLSI[a] (number of isolates)			
Antimicrobial (oral)	**All (3,216)**	**ESBL (541)**	**MDR (317)**	**CRE (57)**
Ceftibuten-avibactam	[99.6 / 98.4][b]	[98.3 / 97.6][b]	[95.6 / 92.1][b]	[78.9 / 73.7][b]
Ceftibuten	89.3[c]	56.7[c]	41.3[c]	19.3[c]
Cefuroxime	61.2[d]	1.1[d]	0.0[d]	0.0[d]
Cefazolin	66.6[c]	0.7[c]	0.0[c]	0.0[c]
Cephalexin	66.6[c]	0.7[c]	0.0[c]	0.0[c]
Levofloxacin	75.4	24.6	11.7	15.8
Ciprofloxacin	72.9	17.7	4.7	14.0
Nitrofurantoin	67.7	63.0	46.1	15.8
TMP-SMX	73.4	32.0	25.6	24.6
Antimicrobial (IV)				
Ceftazidime-avibactam	99.6	98.2	95.6	77.2
Ceftolozane-tazobactam	94.1	82.2	63.6	3.5
Piperacillin-tazobactam	87.7	62.7	32.2	1.8
Ceftriaxone	79.0	6.1	4.4	3.5
Meropenem	98.2	91.3	82.0	3.5
Gentamicin	90.6	66.7	41.6	66.7
Amikacin	99.1	95.6	91.8	71.9
Colistin	85.6[c]	90.7[c]	83.6[c]	70.2[c]

[a] Criteria as published by CLSI (2022).
[b] Values in brackets indicate percentage inhibited at ≤8 / ≤1 mg/L for comparison.
[c] Using uncomplicated urinary tract infection only breakpoints.
[d] Using oral breakpoints.
Abbreviations: TMP-SMX, trimethoprim-sulfamethoxazole; MDR, multidrug-resistant; ESBL, extended-spectrum β-lactamase phenotype; CRE, carbapenem-resistant Enterobacterales.

Conclusions

- Ceftibuten-avibactam was highly active against a large collection of contemporary Enterobacterales isolated from patients with cUTIs and exhibited a similar spectrum to ceftazidime-avibactam.
- Ceftibuten-avibactam retained strong activity against CRE, isolates with an MDR and/or ESBL phenotype, and isolates nonsusceptible to levofloxacin, nitrofurantoin, and/or TMP-SMX.
- Ceftibuten-avibactam may represent a valuable option for oral treatment of cUTI caused by resistant Enterobacterales.

Funding

This study was supported by Pfizer Inc. Helio S. Sader, Cecilia G. Carvalhaes, Michael D. Huband, Rodrigo E. Mendes, and Mariana Castanheira are employees of JMI Laboratories, which was a paid consultant to Pfizer in connection with the development of this poster.

Acknowledgments

The authors thank all participants of the SENTRY Antimicrobial Surveillance Program for their work in providing isolates. Editorial support was provided by Amy Chen at JMI Laboratories and was funded by Pfizer.

References

CLSI. 2022. M100Ed32. Performance standards for antimicrobial susceptibility testing: 32nd informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

Magiorakos AP, Srinivasan A, Carey RB, et al. 2012. Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. Clin. Microbiol. Infect. 18:268-281.

Sanchez GV, Babiker A, Master RN, et al. Antibiotic resistance among urinary isolates from female outpatients in the United States in 2003 and 2012. Antimicrob. Agents Chemother. 2016, 60 (5), 2680-3.

Simmering J, Tang F, Cavanaugh J, et al. The increase in hospitalizations for urinary tract infections and the associated costs in the United States, 1998–2011. Open Forum Infect. Dis. 2017, 4 (1), ofw281.

US FDA 2016. FDA Drug Safety Communication: FDA advises restricting fluoroquinolone antibiotic use for certain uncomplicated infections; warns about disabling side effects that can occur together. Available at: https://www.fda.gov/drugs/drug-safety-and-availability/fda-drug-safety-communication-fda-advises-restricting-fluoroquinolone-antibiotic-use-certain#:~:text=An%20FDA%20safety%20review%20has,nerves%2C%20and%20central%20nervous%20system. Accessed 2022 July 7.

Wiseman LR, Balfour JA, Ceftibuten. A review of its antibacterial activity, pharmacokinetic properties and clinical efficacy. Drugs 1994, 47 (5), 784-808.

Contact

Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ASM-Microbe19-plazomicin-cUTI.pdf

Charges may apply.
No personal information is stored.

IDWeek 2022, October 19–23, 2022, Washington, D.C.