IDWeek 2022 | Poster #P1677

In vitro Activity of SPR206 and Comparator Compounds against Enterobacterales Isolates Responsible for Infections in United States Hospitals

Rodrigo E. Mendes[1], Helio S. Sader[1], S.J. Ryan Arends[1], Nicole Cotroneo[2], Ian A. Critchley[2], Mariana Castanheira[1]

[1] JMI Laboratories, North Liberty, Iowa, USA; [2] Spero Therapeutics, Cambridge, MA, USA

Introduction

- The proportion of isolates producing extended-spectrum β-lactamases (ESBLs) have increased in both hospital and nosocomial settings in the USA.
 - These pathogens are responsible for approximately 197,400 cases and 9,100 deaths per year.
 - In addition, this increased frequency challenges the empiric treatment of serious infections and may promote the use of more potent antimicrobial agents, including carbapenems.
- This scenario helped potentialize the emergence and dissemination of Gram-negative multidrug-resistant (MDR) pathogens in recent decades, including carbapenem-resistant Enterobacterales, where treatment options are often not available.
- SPR206 is a next generation polymyxin under clinical development to treat pneumonia, bloodstream, and urinary tract infections caused by Gram-negative MDR pathogens.
- The *in vitro* activity of SPR206 and comparators was monitored against Gram-negative pathogens causing infection in US and European hospitals during 2021 as part of the SENTRY Antimicrobial Surveillance Program.
- This study reports the activity of SPR206 against Enterobacterales from US hospitals.

Materials and Methods

Bacterial organisms

- This study comprised a collection of 1,614 Enterobacterales collected from various clinical specimens from patients hospitalized in 30 medical centers in 9 US Census Divisions during 2021. Only consecutive isolates (1 per patient infection episode) responsible for documented infections according to local criteria were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories (North Liberty, IA, USA) and contained cation-adjusted Mueller-Hinton broth as per the CLSI guidelines.
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI–recommended quality control reference strains. MIC interpretations were performed using CLSI breakpoints for comparators. A susceptible breakpoint of ≤2 mg/L was used for SPR206 for comparison purposes.

Subset definitions

- ESBL producers were presumptively defined as *Escherichia coli*, *Klebsiella pneumoniae*, and *Proteus mirabilis* with ceftriaxone, ceftazidime, or aztreonam MICs ≥2 mg/L.
- MDR was defined as any isolate resistant to ≥3 classes of antibiotics, whereas an extensively drug-resistant phenotype as defined as any isolate susceptible to ≤2 classes of antibiotics.
- CRE was defined as any Enterobacterales displaying MIC values ≥2 mg/L (CLSI) for imipenem (*P. mirabilis, P. penneri*, and indole-positive Proteeae were excluded) or meropenem.
- The CRE definition was also used to define isolates as not susceptible to carbapenems (CLSI).

Results

- *E. coli* (*n*=425) and *K. pneumoniae* (*n*=425) were the most common pathogens included in this surveillance study, followed by *E. cloacae* (*n*=215), *Citrobacter* spp. (*n*=120), *K. oxytoca* (*n*=110), *S. marcescens* (*n*=104), *K. aerogenes* (*n*=65), *P. mirabilis* (*n*=60), *M. morganii* (*n*=60), and 11 other species (*n*=30).
- Among *E. coli, K. pneumoniae*, and *P. mirabilis*, 170 (18.7%) isolates met the MIC criteria for the presumptive production of ESBL, whereas 128 (9.4%) and 21 (1.5%) isolates were categorized as MDR and XDR, respectively (Table 1).
 - A total of 39 (2.9%) isolates were not susceptible to the carbapenems, imipenem, and/or meropenem (Table 1).
- Overall, SPR206 and colistin had MIC_{50} values of 0.06 and 0.25 mg/L, respectively, against all Enterobacterales, and against Enterobacterales excluding those that are intrinsically resistant to polymyxins (Figure 1).
 - *Morganella* spp., *Proteus* spp., *Providencia* spp., and *Serratia* spp. that were intrinsically resistant to polymyxins (MIC, ≥8 mg/L) also had an elevated MIC against SPR206 (MIC, ≥8 mg/L).
 - Excluding these organisms, SPR206 ($MIC_{50/90}$, 0.06/0.12 mg/L) and meropenem ($MIC_{50/90}$, 0.03/0.06 mg/L) showed the lowest MIC against this Enterobacterales subset, followed by colistin ($MIC_{50/90}$, 0.25/0.25 mg/L) and ceftazidime-avibactam ($MIC_{50/90}$, 0.12/0.5 mg/L).
- SPR206 displayed MIC_{50} and MIC_{90} results of 0.06 and 0.12 mg/L, respectively, against both *E. coli* and *K. pneumoniae* (Table 1).
- Similar MIC_{50} and MIC_{90} values of 0.06 and 0.12–0.5 mg/L, respectively, were obtained for SPR206 against the ESBL, MDR, XDR, and CRE resistant subsets.
 - Colistin ($MIC_{50/90}$, 0.25/0.25–0.5 mg/L) was also active against these subsets, inhibiting 94.9–97.6% of isolates at ≤2 mg/L, whereas ceftazidime-avibactam ($MIC_{50/90}$, 0.25/1–2 mg/L) showed susceptibility rates of 96.1–98.2% against the ESBL and MDR sets (Table 2).

Figure 1. Cumulative MIC distribution of SPR206 and colistin against US Enterobacterales and resistant subsets

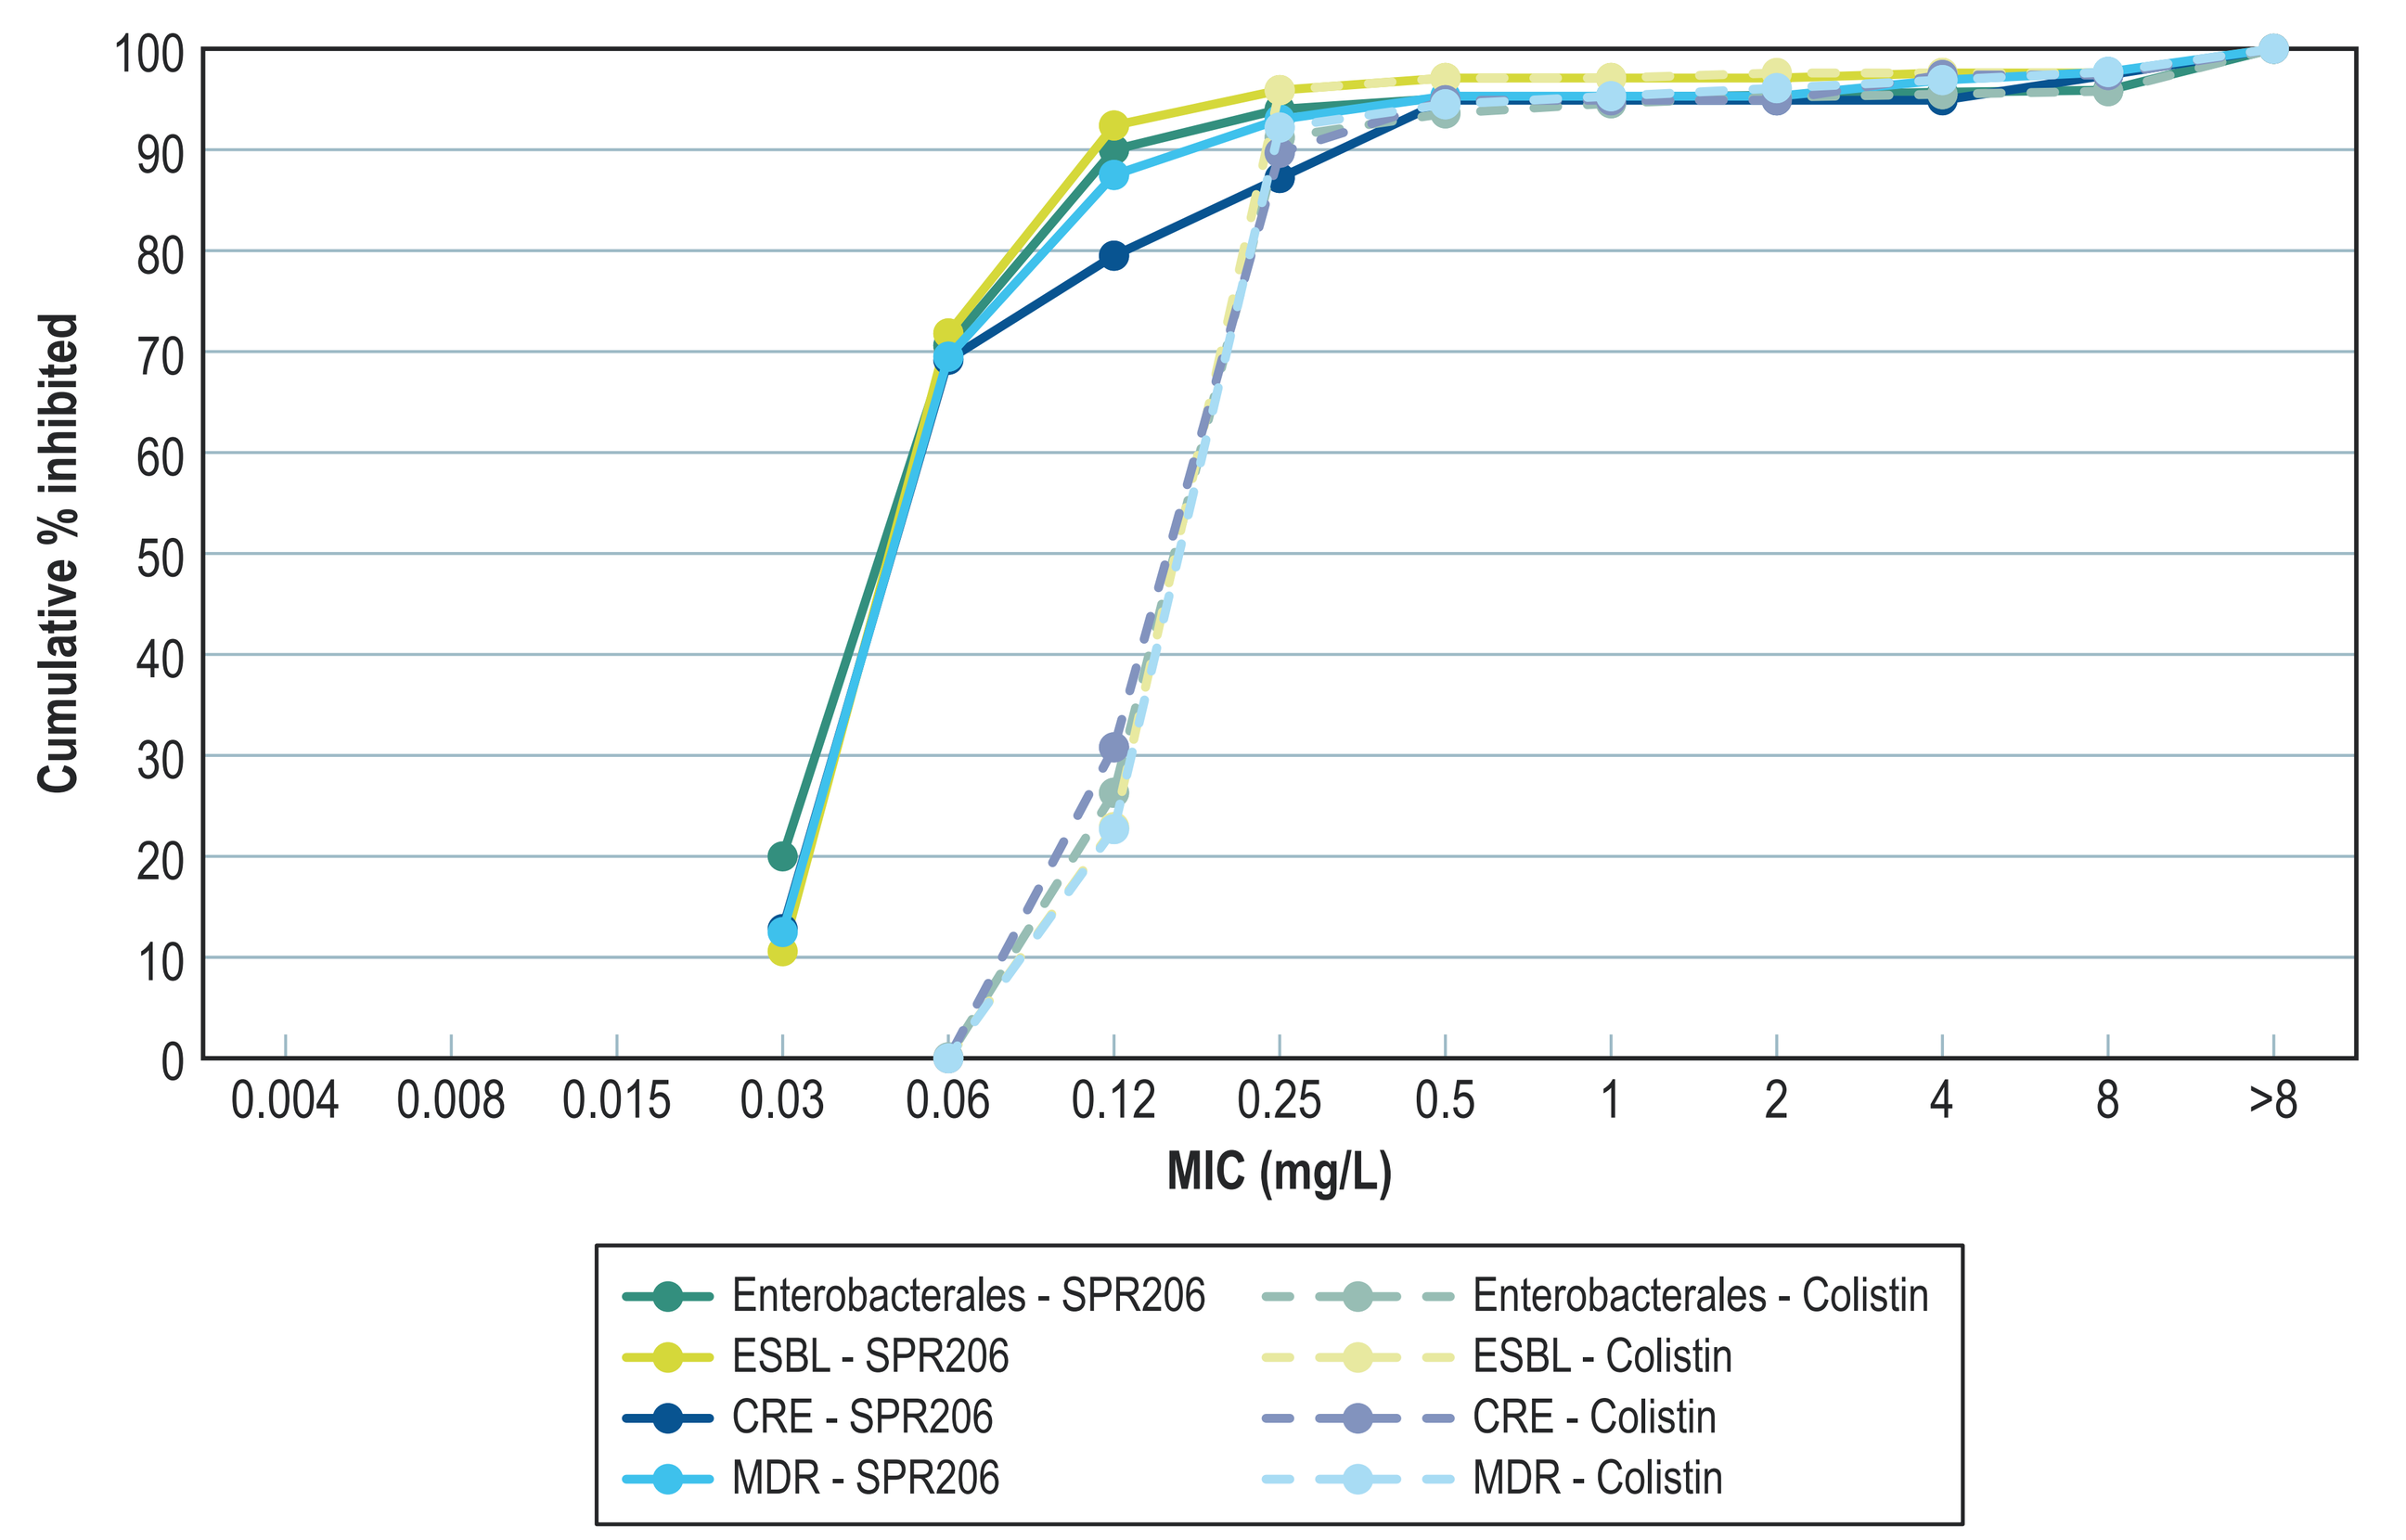
Cumulative % inhibited
100
90
80
70
60
50
40
30
20
10
0
0.004
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
>8
MIC (mg/L)
Enterobacterales - SPR206
ESBL - SPR206
CRE - SPR206
MDR - SPR206
Enterobacterales - Colistin
ESBL - Colistin
CRE - Colistin
MDR - Colistin

Table 1. MIC distribution of SPR206 obtained against US Enterobacterales and resistant subsets

Organism/ Group (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (mg/L) of:													MIC_{50}	MIC_{90}
	≤0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	>64		
All Enterobacterales (1,614)	270 (16.7)	694 (59.7)	265 (76.1)	55 (79.6)	15 (80.5)	2 (80.6)	2 (80.7)	4 (81.0)	6 (81.4)	11 (82.0)	11 (82.7)	11 (83.4)	268 (100)	0.06	>64
Enterobacterales (1,366)[a]	270 (19.8)	694 (70.6)	265 (90.0)	55 (94.0)	15 (95.1)	2 (95.2)	2 (95.4)	4 (95.7)	3 (95.9)	6 (96.3)	6 (96.8)	6 (97.2)	38 (100)	0.06	0.25
E. coli (425)	108 (25.4)	263 (87.3)	49 (98.8)	5 (100.0)										0.06	0.12
K. pneumoniae (425)	48 (11.3)	213 (61.4)	124 (90.6)	22 (95.8)	4 (96.7)	2 (97.2)	0 (97.2)	1 (97.4)	1 (97.6)	5 (98.8)	3 (99.5)	1 (99.8)	1 (100)	0.06	0.12
ESBL (170)[b]	18 (10.6)	104 (71.8)	35 (92.4)	6 (95.9)	2 (97.1)	0 (97.1)	0 (97.1)	1 (97.6)	0 (97.6)	3 (99.4)	1 (100.0)			0.06	0.12
MDR (128)[d]	16 (12.5)	73 (69.5)	23 (87.5)	7 (93.0)	3 (95.3)	0 (95.3)	0 (95.3)	2 (96.9)	1 (97.7)	1 (98.4)	0 (98.4)	0 (98.4)	2 (100)	0.06	0.25
CRE (39)[c]	5 (12.8)	22 (69.2)	4 (79.5)	3 (87.2)	3 (94.9)	0 (94.9)	0 (94.9)	0 (94.9)	1 (97.4)	1 (100.0)				0.06	0.5
XDR (21)[e]	1 (4.8)	14 (71.4)	3 (85.7)	1 (90.5)	2 (100.0)									0.06	0.25

[a] Excludes intrinsically resistant isolates, such as *Morganella* spp., *Proteus* spp., *Providencia* spp., and *Serratia* spp.
[b] Includes 78 *E. coli* and 92 *K. pneumoniae* isolates that met the MIC criteria for the presumptive production of ESBL (i.e., ceftriaxone, ceftazidime, or aztreonam MIC values ≥2 mg/L).
[c] Enterobacterales that were not susceptible to carbapenems (imipenem and/or meropenem), among which 31 were carbapenem-resistant isolates. Includes 1 *C. freundii* species complex, 7 *E. cloacae* species complex, 1 *E. coli*, 8 *K. aerogenes*, 2 *K. oxytoca*, and 20 *K. pneumoniae*.
[d] MDR, multidrug-resistant (resistant to ≥3 classes) isolates; includes 2 *C. freundii* species complex, 1 *C. koseri*, 12 *E. cloacae* species complex, 36 *E. coli*, 1 *H. alvei*, 9 *K. aerogenes*, 9 *K. oxytoca*, and 58 *K. pneumoniae*.
[e] XDR, extensively drug-resistant (susceptible to ≤2 classes); includes 3 *E. cloacae* species complex, 1 *E. coli*, 1 *K. aerogenes*, 1 *K. oxytoca*, 15 *K. pneumoniae*.

Table 2. Antimicrobial activity of SPR206 and comparator agents tested against US Enterobacterales and resistant subsets

Antimicrobial agent	MIC (mg/L)			CLSI[a]		
	50%	90%	Range	%S	%I	%R
Enterobacterales (1,366)[a]						
SPR206[a]	0.06	0.12	≤0.03 to >64	95.4		
Colistin	0.25	0.25	≤0.06 to >8		95.2	4.8
Aztreonam	0.12	>16	≤0.03 to >16	79.4	1.4	19.2
Meropenem	0.03	0.06	≤0.015 to >32	97.4	0.7	1.9
Imipenem	≤0.12	0.5	≤0.12 to >8	97.4	0.5	2.1
Ceftazidime	0.25	>32	0.03 to >32	80.1	1.1	18.8
Ceftazidime-avibactam	0.12	0.5	≤0.015 to >32	99.6		0.4
Ceftriaxone	≤0.06	>8	≤0.06 to >8	77.4	1.0	21.7
Piperacillin-tazobactam	2	64	0.12 to >128	81.3	5.2	13.5
Amikacin	2	4	≤0.25 to >32	99.3	0.3	0.4
Tobramycin	0.5	4	≤0.12 to >16	90.6	2.7	6.7
Tigecycline	0.25	0.5	≤0.06 to 8			
Levofloxacin	0.06	8	≤0.015 to >32	84.4	2.6	13.0
Trimethoprim-sulfamethoxazole	≤0.12	>4	≤0.12 to >4	81.7		18.3
Ceftolozane-tazobactam	0.25	2	≤0.12 to >16	90.3	2.2	7.5
ESBL (170)[b]						
SPR206[a]	0.06	0.12	≤0.03 to 32	97.1		
Colistin	0.25	0.25	0.12 to >8		97.6	2.4
Aztreonam	>16	>16	0.25 to >16	11.8	5.9	82.4
Meropenem	0.03	4	≤0.015 to >32	87.6	1.8	10.6
Imipenem	≤0.12	4	≤0.12 to >8	88.8	0.6	10.6
Ceftazidime	32	>32	0.25 to >32	13.5	7.1	79.4
Ceftazidime-avibactam	0.25	1	≤0.015 to >32	98.2		1.8
Ceftriaxone	>8	>8	0.25 to >8	5.9	2.4	91.8
Piperacillin-tazobactam	8	>128	1 to >128	52.9	14.1	32.9
Amikacin	4	8	≤0.25 to >32	94.1	2.4	3.5
Tobramycin	4	>16	≤0.12 to >16	50.0	11.8	38.2
Tigecycline	0.25	1	0.12 to 8			
Levofloxacin	8	32	0.03 to >32	28.2	10.0	61.8
Trimethoprim-sulfamethoxazole	>4	>4	≤0.12 to >4	32.4		67.6
Ceftolozane-tazobactam	1	>16	≤0.12 to >16	76.3	5.9	17.8

Antimicrobial agent	MIC (mg/L)			CLSI[a]		
	50%	90%	Range	%S	%I	%R
CRE (39)[b]						
SPR206[a]	0.06	0.5	≤0.03 to 16	94.9		
Colistin	0.25	0.5	0.12 to >8		94.9	5.1
Aztreonam	>16	>16	0.25 to >16	7.7	0.0	92.3
Meropenem	4	>32	0.06 to >32	7.7	25.6	66.7
Imipenem	8	>8	0.5 to >8	7.7	17.9	74.4
Ceftazidime	>32	>32	0.5 to >32	7.7	2.6	89.7
Ceftazidime-avibactam	1	>32	≤0.015 to >32	87.2		12.8
Ceftriaxone	>8	>8	0.12 to >8	5.1	0.0	94.9
Piperacillin-tazobactam	>128	>128	4 to >128	5.1	2.6	92.3
Amikacin	4	>32	0.5 to >32	82.1	5.1	12.8
Tobramycin	8	>16	0.25 to >16	43.6	7.7	48.7
Tigecycline	0.5	4	0.12 to 4			
Levofloxacin	2	>32	≤0.015 to >32	35.9	12.8	51.3
Trimethoprim-sulfamethoxazole	4	>4	≤0.12 to >4	48.7		51.3
Ceftolozane-tazobactam	>16	>16	0.5 to >16	7.7	2.6	89.7
MDR (128)[b]						
SPR206[a]	0.06	0.25	≤0.03 to >64	95.3		
Colistin	0.25	0.25	0.12 to >8		96.1	3.9
Aztreonam	>16	>16	0.06 to >16	4.7	2.3	93.0
Meropenem	0.03	32	≤0.015 to >32	71.9	7.8	20.3
Imipenem	≤0.12	>8	≤0.12 to >8	73.4	3.9	22.7
Ceftazidime	>32	>32	0.25 to >32	7.0	1.6	91.4
Ceftazidime-avibactam	0.25	2	≤0.015 to >32	96.1		3.9
Ceftriaxone	>8	>8	≤0.06 to >8	1.6	0.8	97.7
Piperacillin-tazobactam	32	>128	1 to >128	18.8	21.9	59.4
Amikacin	4	16	≤0.25 to >32	92.2	3.1	4.7
Tobramycin	16	>16	≤0.12 to >16	24.2	17.2	58.6
Tigecycline	0.5	2	0.12 to 8			
Levofloxacin	8	>32	≤0.015 to >32	21.1	13.3	65.6
Trimethoprim-sulfamethoxazole	>4	>4	≤0.12 to >4	31.2		68.8
Ceftolozane-tazobactam	2	>16	≤0.12 to >16	55.5	7.0	37.5

[a] Criteria as published by CLSI (2021); excludes intrinsically resistant isolates, such as *Morganella* spp., *Proteus* spp., *Providencia* spp., and *Serratia* spp.; Percent susceptible described for SPR206 represents isolates inhibited at MIC of ≤2 mg/L, for comparison purposes.
[b] See footnotes on Table 1 for additional information.

Conclusions

- SPR206 was consistently more potent than its in-class comparator colistin against these pathogens and their resistance subsets causing infections in US hospitals.
 - SPR206 remained *in vitro* active against all resistant subsets, including ESBL, CRE, MDR, and XDR groups, where limited intravenous options were available.
- These data, combined with the favorable safety and tolerability profiles in Phase 1 studies, support the continued clinical advancement for SPR206.

Acknowledgements

This research and poster presentation were sponsored by Spero Therapeutics, which was supported by the Office of the Assistant Secretary of Defense for Health Affairs through the Joint Warfighter Medical Research Program under Award No. W81XWH-19-1-0295.

References

1. Bruss J, Lister T, Gupta VK, Stone E, Morelli L, Lei Y, Melnick D (2021). Single- and multiple-ascending-dose study of the safety, tolerability, and pharmacokinetics of the polymyxin derivative SPR206. *Antimicrob. Agents Chemother*. 65 (10): e0073921.
2. Brown P, Abbott E, Abdulle O, Boakes S, Coleman S, Divall N, Duperchy E, Moss S, Rivers D, Simonovic M, Singh J, Stanway S, Wilson A, Dawson MJ (2019). Design of next generation polymyxins with lower toxicity: The discovery of SPR206. *ACS Infect Dis*. 5 (10): 1645–1656.
3. Clinical and Laboratory Standards Institute (2018). Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. M07 11th Edition. Wayne, PA, USA.
4. Clinical and Laboratory Standards Institute (2022). Performance standards for antimicrobial susceptibility testing. M100 32nd Edition. Wayne, PA, USA.

Contact

Rodrigo E. Mendes, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2022_SPR206VsEnteros.pdf

No personal information is stored.

IDWeek 2022, October 19–23, 2022, Washington, D.C.